Differentiation of Isobaric and Isomeric Fentanyl Analogs by Gas Chromatography-Mass Spectrometry (GC-MS)

Jianmei Liu and Roxanne E. Franckowski, MS
Cayman Chemical, Ann Arbor, Michigan www.caymanchem.com

Abstract

The structural similarities and isomeric nature of fentanyl analogs makes their differentiation a big challenge. The combination of mass spectrometry (MS) with gas chromatography (GC) is a promising way to solve the problem. In the present study, 60 fentanyl analogs, including structural and geometric isomers, were analyzed by GC-MS. An extracted ion chromatogram (EIC) function was used to select key fragments of the fentanyl analogs. Relative retention time (RRT) was used to minimize the impact of retention time variation. Oven temperature was determined to be critical to achieving successful separation. By slowing down the rate of the oven temperature program, complete chromatographic separation and baseline resolution of more than 1.5 was achieved. In this study, the effects of tuning type were also investigated. Tuning of the mass spectrometer affected the ratio of the characteristic fragment ions. However, isomers could not be differentiated by simply altering tuning type as they would likely have similar fragments with the same tuning method.

Introduction

Fentanyl, a powerful pharmaceutical-grade opioid, has been used for decades to treat pain. Numerous fentanyl analogs began to emerge in the illicit market in 2015. To date, a wide variety of fentanyl analogs, such as cyclopropyl fentanyl and 3-methylfentanyl, have been identified by forensic practitioners. Isobaric and isomeric analogs of fentanyl pose challenges for identification and differentiation of these compounds in forensic casework. There have been a number of publications on the differentiation of the fentanyl analogs[1-3]; however, this poster and supplemental material will provide gas chromatography (GC) separation conditions for 60 fentanyl analogs, reintroduce forensic practitioners to the usefulness of extracted ion chromatograms (EICs), and explore the effects of different tuning conditions on isobaric compounds.

Methods

Fentanyl (hydrochloride) (CRM) and all fentanyl analogs (provided as neat materials) used in the study are Cayman products. Single component solutions for each fentanyl analog were prepared by dissolving 1 mg of neat material in 1 ml of HPLC-grade methanol (EMD Millipore). Twenty multicomponent mixtures were prepared by transferring 200 µl aliquots of each single component solution with a 200 µl aliquot of internal standard into a vial and dried under nitrogen. The multicomponent mixtures were reconstituted with 200 µl of methanol for a final nominal concentration of 1.0 mg/ml per component. The multicomponent solutions were transferred to autosampler vials with insert prior to injection. The 20 multicomponent mixtures were used in the GC separation study.

A multicomponent solution containing all 60 components was prepared by transferring 200 µl aliquots of each single component solution into a vial and dried under nitrogen. The 60-component mixture was reconstituted with 200 µl of methanol for a final nominal concentration of 1.0 mg/ml per component. The 60-component mixture was used in the EIC study.

A 1.0 µl injection of each solution was analyzed using the instrument and parameters listed in **Table 1**. A Standard Spectra Autotune (S-tune) was performed prior to sample analysis in the study.

Table 1. GC-MS method conditions

Instrument	Agilent 6890 Gas Chromatograph equipped with an Agilent 5973 Mass Selective Detector	
Column	Restek, Rtx-5 MS, 30 m × 0.32 mm I.D., 0.5 µm film thickness (Phase composition: Crossbond 5% diphenyl / 95% dimethyl polysiloxane; similar column: DB-5MS)	
Injector Temperature	300°C	
Oven Temperature	*Method 1:* 50°C for 1 minute, 30°C/minute to 300°C. Total run time: 15 minutes *Method 2:* 240°C for 1 minute, 1°C/minute to 260°C, 30°C/minute to 300°C. Total run time: 26 minutes *Method 3:* 50°C for 1 minute, 30°C/minute to 300°C. Total run time: 30 minutes	
Carrier Gas	Helium at 2.0 ml/minute, split ratio = 15:1	
MS Settings	*Transfer line temperature:*	300°C
	MS Source:	230°C
	MS Quad:	150°C
	Scan Range:	40-600 *m/z*
	Electron Ionization:	70eV
S-tune Parameters	*Target Tune Masses:*	69, 219, 502
	69 Abundance Target, Counts:	8,000,000
	Mass 219 Target %:	55
	Mass 502 Target %:	2.5

Result and Discussion

GC Separation

To achieve baseline separation on the GC, multicomponent mixtures of the fentanyl analogs were separated into 20 isomer groups. Three methods were developed for separating each isomer group. Baseline resolution (≥1.5) was obtained for all the isomer groups as illustrated on the following tables. For brevity, representative groups for each method were provided in **Tables 2-4** (see supplemental material for complete list). Relative retention time (RRT) was used to minimize the impact of retention time variation. RRT is expressed as the ratio of retention time of a compound to the internal standard or reference. For this study, fentanyl (hydrochloride) (CRM) was introduced as an internal standard and mixed in all samples. The combination of RRT and MS was used to identify each component.

Ask to see our Supplemental Data

- RRT, resolution, *m/z*, and TIC for 60 fentanyl analogs
- Compare GC separation using three different oven temperature ramps
- This data highlights some of the most common fentanyl analogs

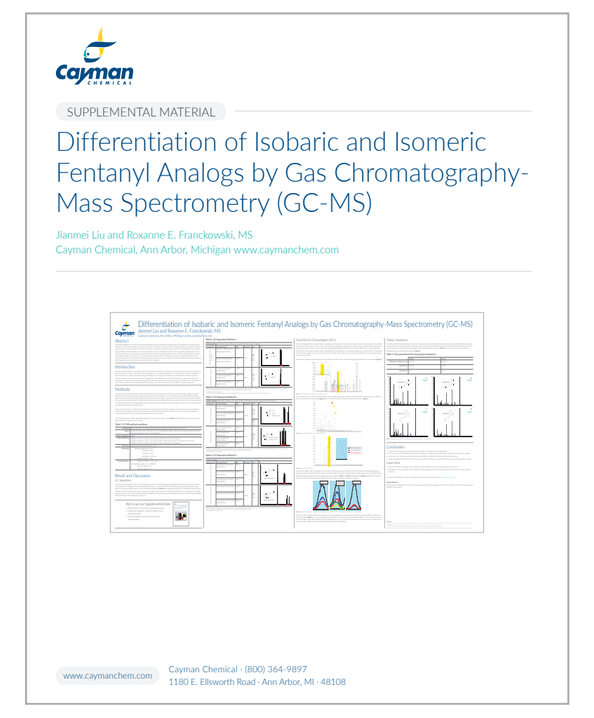
Differentiation of Isobaric and Isomeric Fentanyl Analogs by Gas Chromatography-Mass Spectrometry (GC-MS)

Table 2. GC Separation Method 1

Method conditions: 50°C for 1 minute, 30°C/minute to 300°C. Total run time: 15 minutes

Group Name	Compound Name	RRT*	Resolution	*m/z***	TIC
Fluoro Methoxyacetyl fentanyl	*meta*-fluoro Methoxyacetyl fentanyl (hydrochloride)	1.025	≥1.96	**279** 236 176	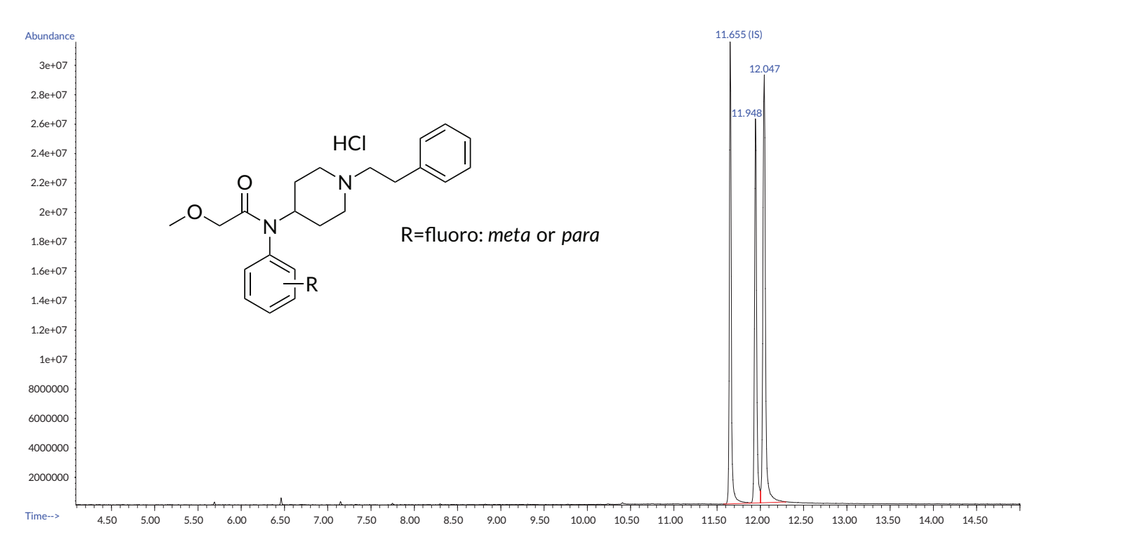
	para-fluoro Methoxyacetyl fentanyl (hydrochloride)	1.034			
Methyl Acetyl fentanyl	2'-methyl Acetyl fentanyl (hydrochloride)	1.000***	≥1.73	**231** 146 188	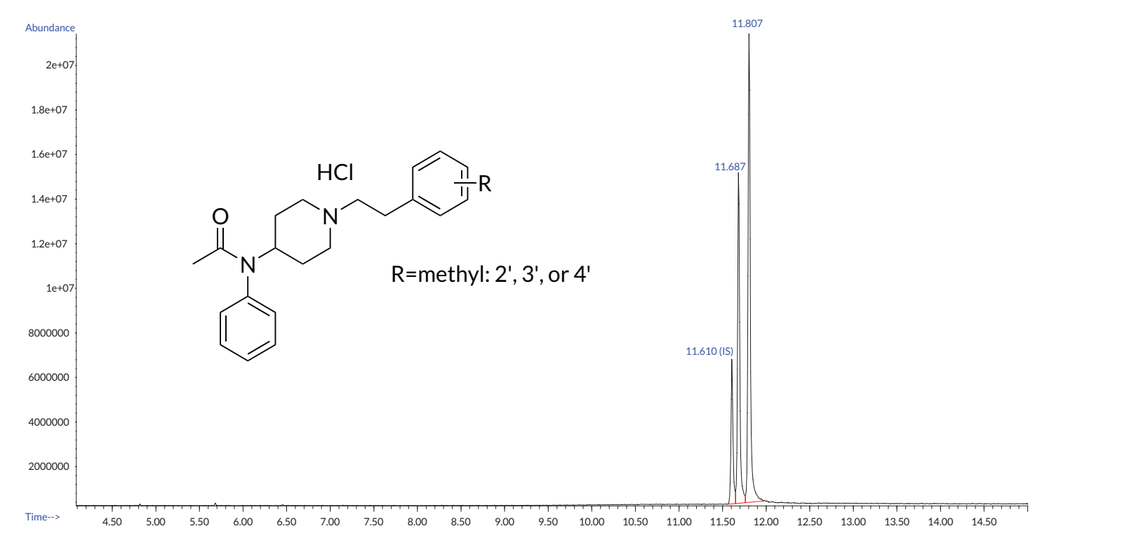
	3'-methyl Acetyl fentanyl (hydrochloride)	1.007			
	4'-methyl Acetyl fentanyl (hydrochloride)	1.017			

HCl
R=fluoro: *meta* or *para*
R=methyl: 2', 3', or 4'

*RRT: Relative retention time (RRT) is the ratio of the retention time of analyte peak relative to that of another used as a reference obtained under identical conditions. RRT = ($T_{analyte}$ / $T_{reference}$) where T = Retention time. Fentanyl (hydrochloride) (CRM) was used as a reference (internal) standard (IS) in the study.
***m/z* in bold represents the base peak.
***2'-methyl Acetyl fentanyl (hydrochloride) and fentanyl (hydrochloride) (CRM) (IS) were coeluted in this condition, therefore, the RRT is 1.000 in the study.

Table 3. GC Separation Method 2

Method conditions: 240°C for 1 minute, 1°C/minute to 260°C, 30°C/minute to 300°C. Total run time: 26 minutes

Group Name	Compound Name	RRT*	Resolution	*m/z***	TIC
Methyl Thiofentanyl	(±)-*trans*-3-methyl Thiofentanyl (hydrochloride)	1.028	≥1.48	**259** 160 203	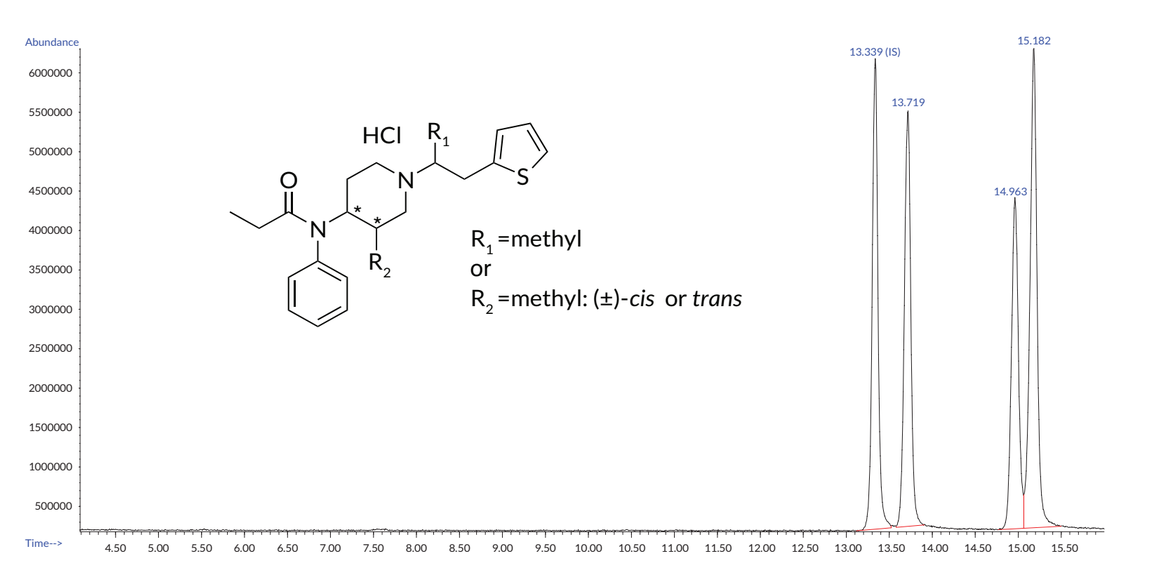
	(±)-*cis*-3-methyl Thiofentanyl (hydrochloride)	1.122			
	α-methyl Thiofentanyl (hydrochloride)	1.138		**259** 110 146	
Methylfentanyl	(±)-*cis*-3-methyl Fentanyl (hydrochloride)	1.084	≥1.95	**259** 160 203	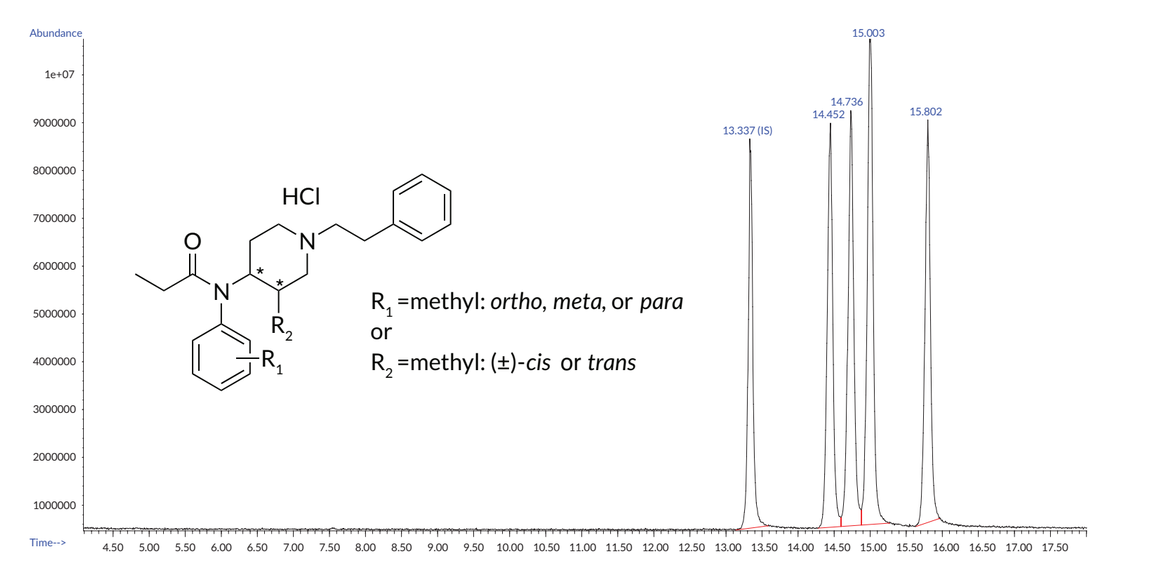
	meta-Methylfentanyl (hydrochloride)	1.105			
	ortho-Methylfentanyl (hydrochloride)	1.125			
	para-Methylfentanyl (hydrochloride)	1.185			

HCl
R1 =methyl or R2 =methyl: (±)-*cis* or *trans*
R1 =methyl: *ortho*, *meta*, or *para* or R2 =methyl: (±)-*cis* or *trans*

*RRT: Relative retention time (RRT) is the ratio of the retention time of analyte peak relative to that of another used as a reference obtained under identical conditions. RRT = ($T_{analyte}$ / $T_{reference}$) where T = Retention time. Fentanyl (hydrochloride) (CRM) was used as a reference (internal) standard (IS) in the study.
***m/z* in bold represents the base peak.

Table 4. GC Separation Method 3

Method conditions: 50°C for 1 minute, 30°C/minute to 300°C. Total run time: 30 minutes

Group Name	Compound Name	RRT*	Resolution	*m/z***	TIC
Methyl Furanyl fentanyl	*meta*-methyl Furanyl fentanyl (hydrochloride)	1.235	≥2.34	**95** 297 254	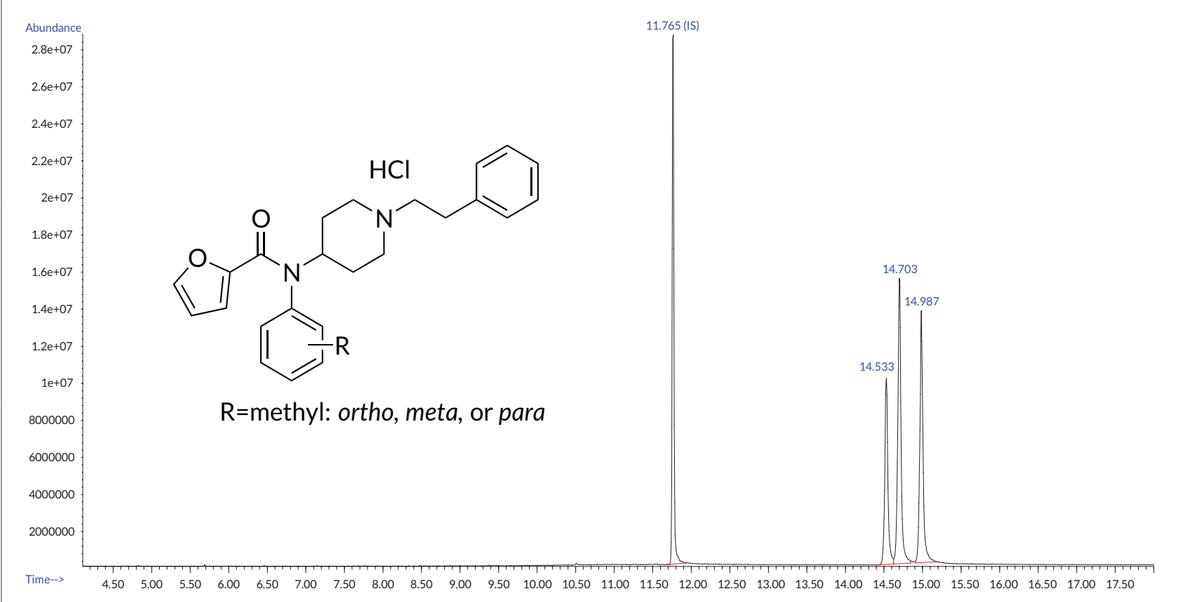
	ortho-methyl Furanyl fentanyl (hydrochloride)	1.250			
	para-methyl Furanyl fentanyl (hydrochloride)	1.274			
Methoxy Furanyl fentanyl	*ortho*-methoxy Furanyl fentanyl	1.345	≥3.08	**313** 95 270	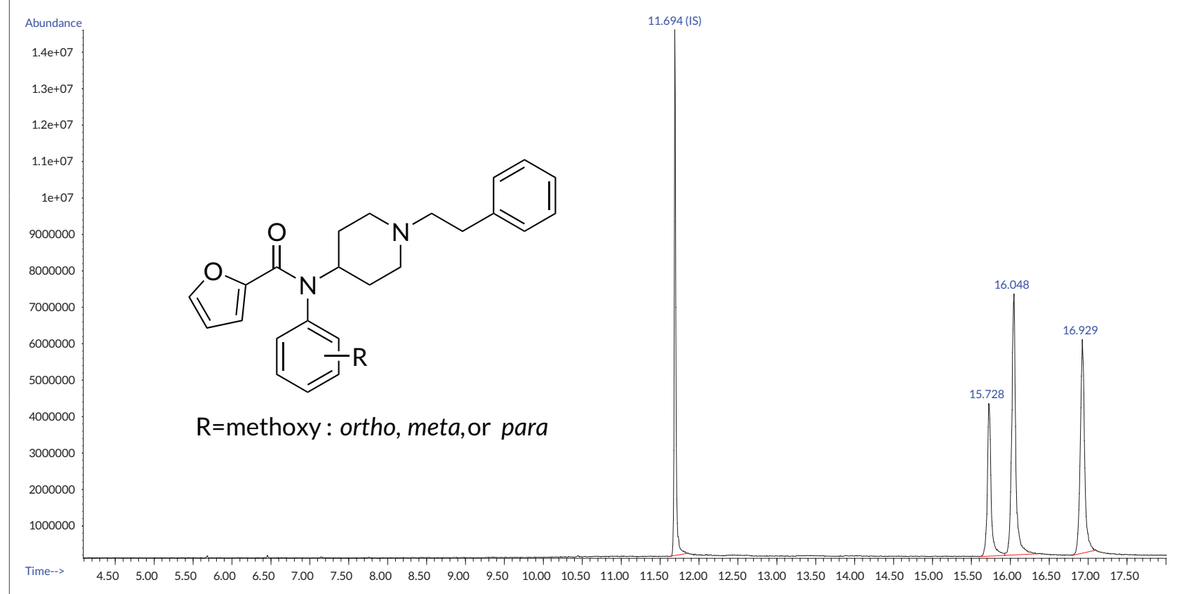
	meta-methoxy Furanyl fentanyl	1.372			
	para-methoxy Furanyl fentanyl (hydrochloride)	1.448			

HCl
R=methyl: *ortho*, *meta*, or *para*
R=methoxy : *ortho*, *meta*,or *para*

*RRT: Relative retention time (RRT) is the ratio of the retention time of analyte peak relative to that of another used as a reference obtained under identical conditions. RRT = ($T_{analyte}$ / $T_{reference}$) where T = Retention time. Fentanyl (hydrochloride) (CRM) was used as a reference (internal) standard (IS) in the study.
***m/z* in bold represents the base peak.

Extracted Ion Chromatograms (EICs)

When chromatographically analyzing mixtures, depending on the instrument conditions and complexity of the mixture, coelution is a possibility. To examine the worst-case scenario, a mixture of the 60 fentanyl analogs was prepared at 1 mg/ml per analyte in methanol and analyzed with Method 2 mentioned in Table 1. Isomers were differentiated using Agilent's Extracted Ion Chromatogram (EIC) function combined with RRT. EIC allows the user to better visualize analytes throughout the chromatographic run by selecting targeted ions from the total ion chromatogram (TIC). RRT of the analyte may be used to further discern the identity of the analyte. An analyte may be identified when key ion fragments were present at a known RRT.

To illustrate EIC, analytes with retention time from 14.839 min to 15.042 min in the TIC of fentanyl analogs standard mixture were examined (**Figure 1**).

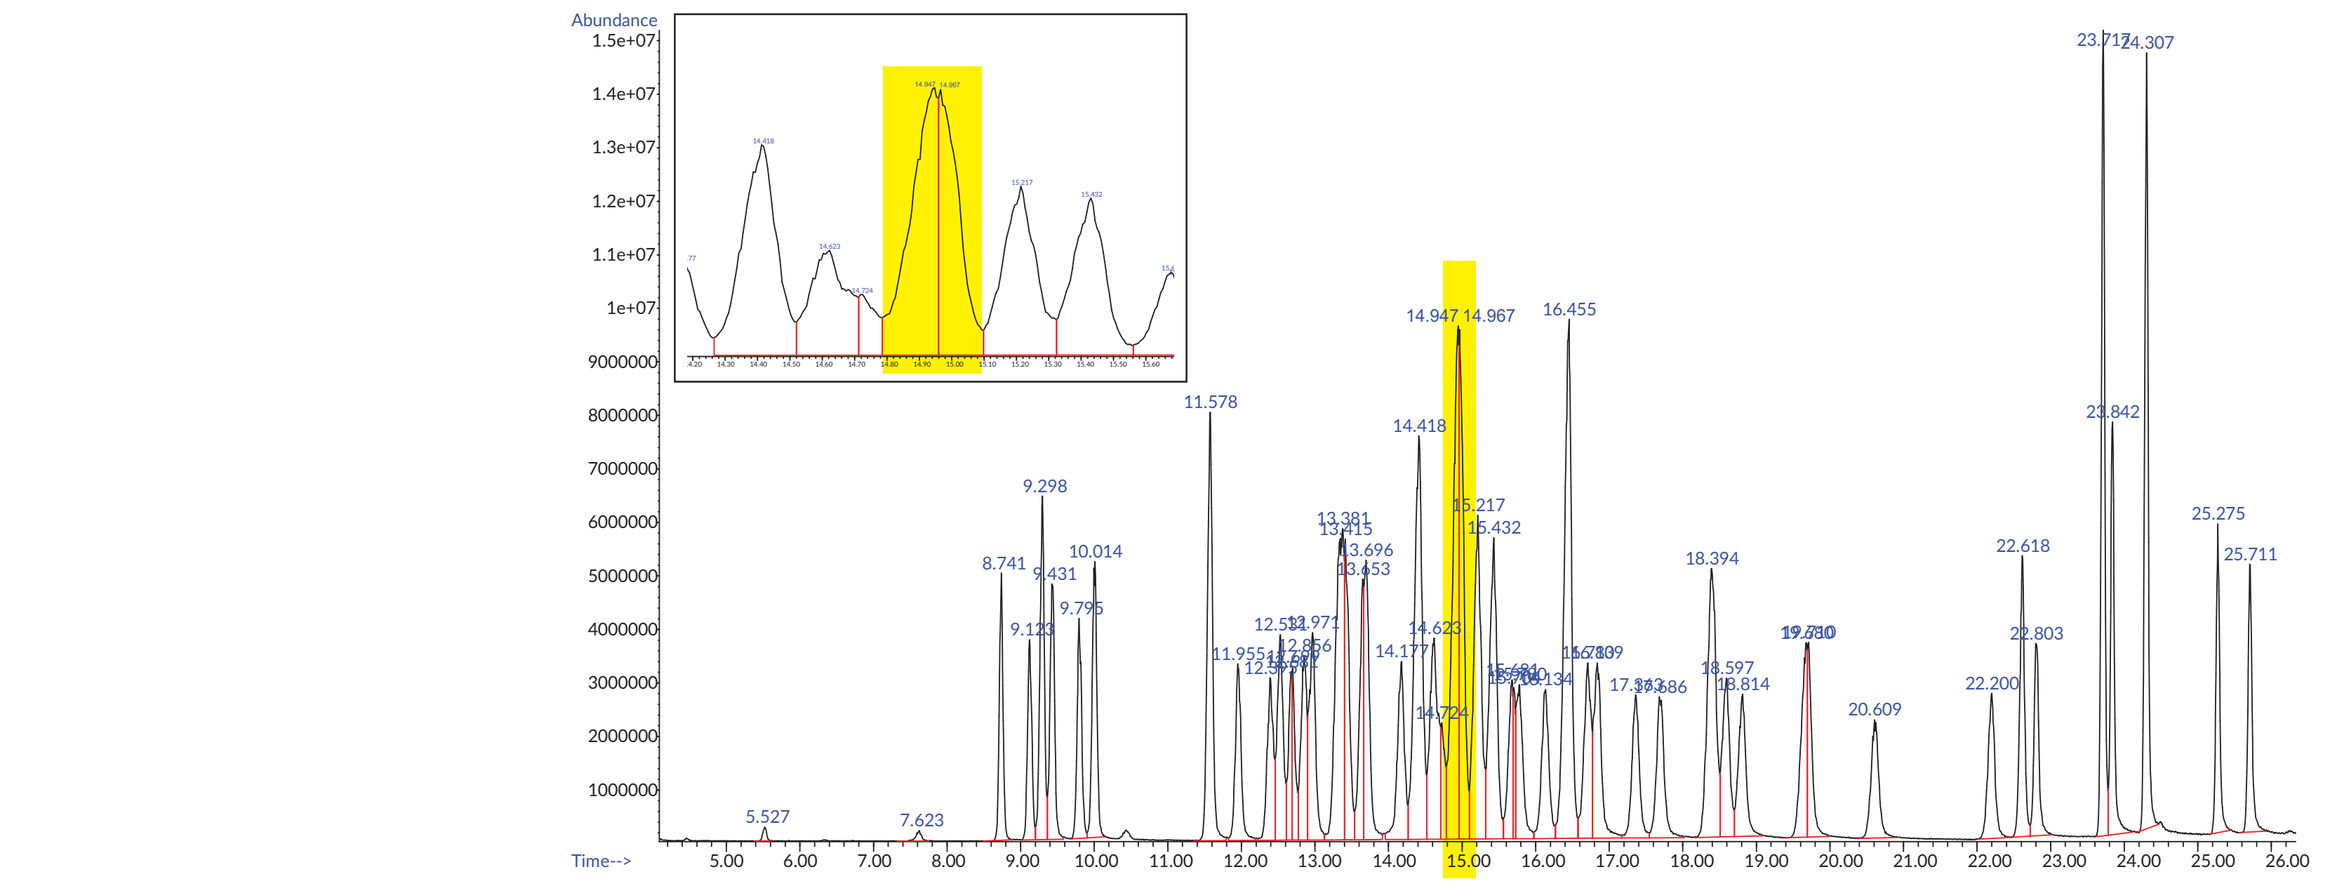

Figure 1. GC-MS chromatogram of 60 fentanyl analogs mixture standard.

TIC was collected in scan mode. Using knowledge of fentanyl analog fragmentation patterns,[4] the high abundance fragments (*m/z* 231, 259, and 279) were suspected to be base peaks (**Figure 2**). After extraction, the EIC of the 231, 259, and 279 are shown in **Figure 3**.

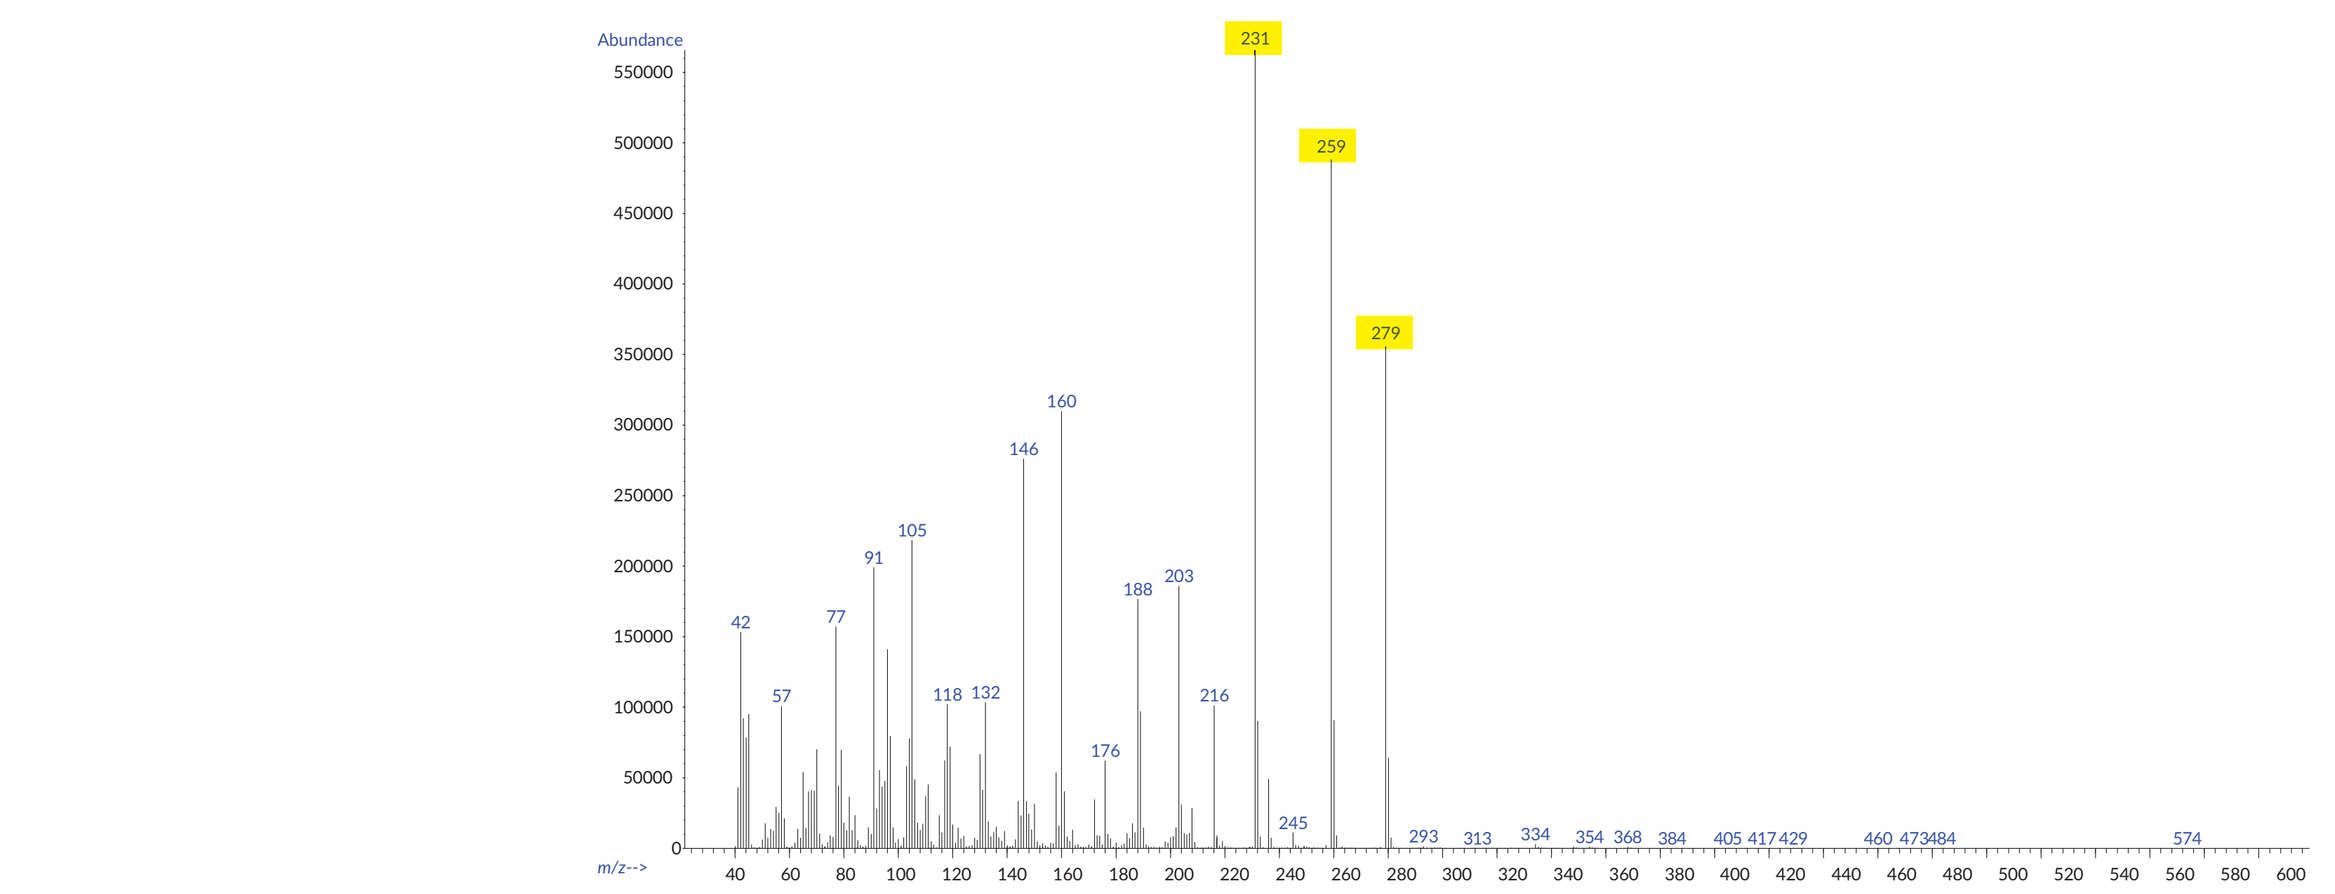

Figure 2. Mass spectrum of TIC averaged from 14.834 to 15.042 min.

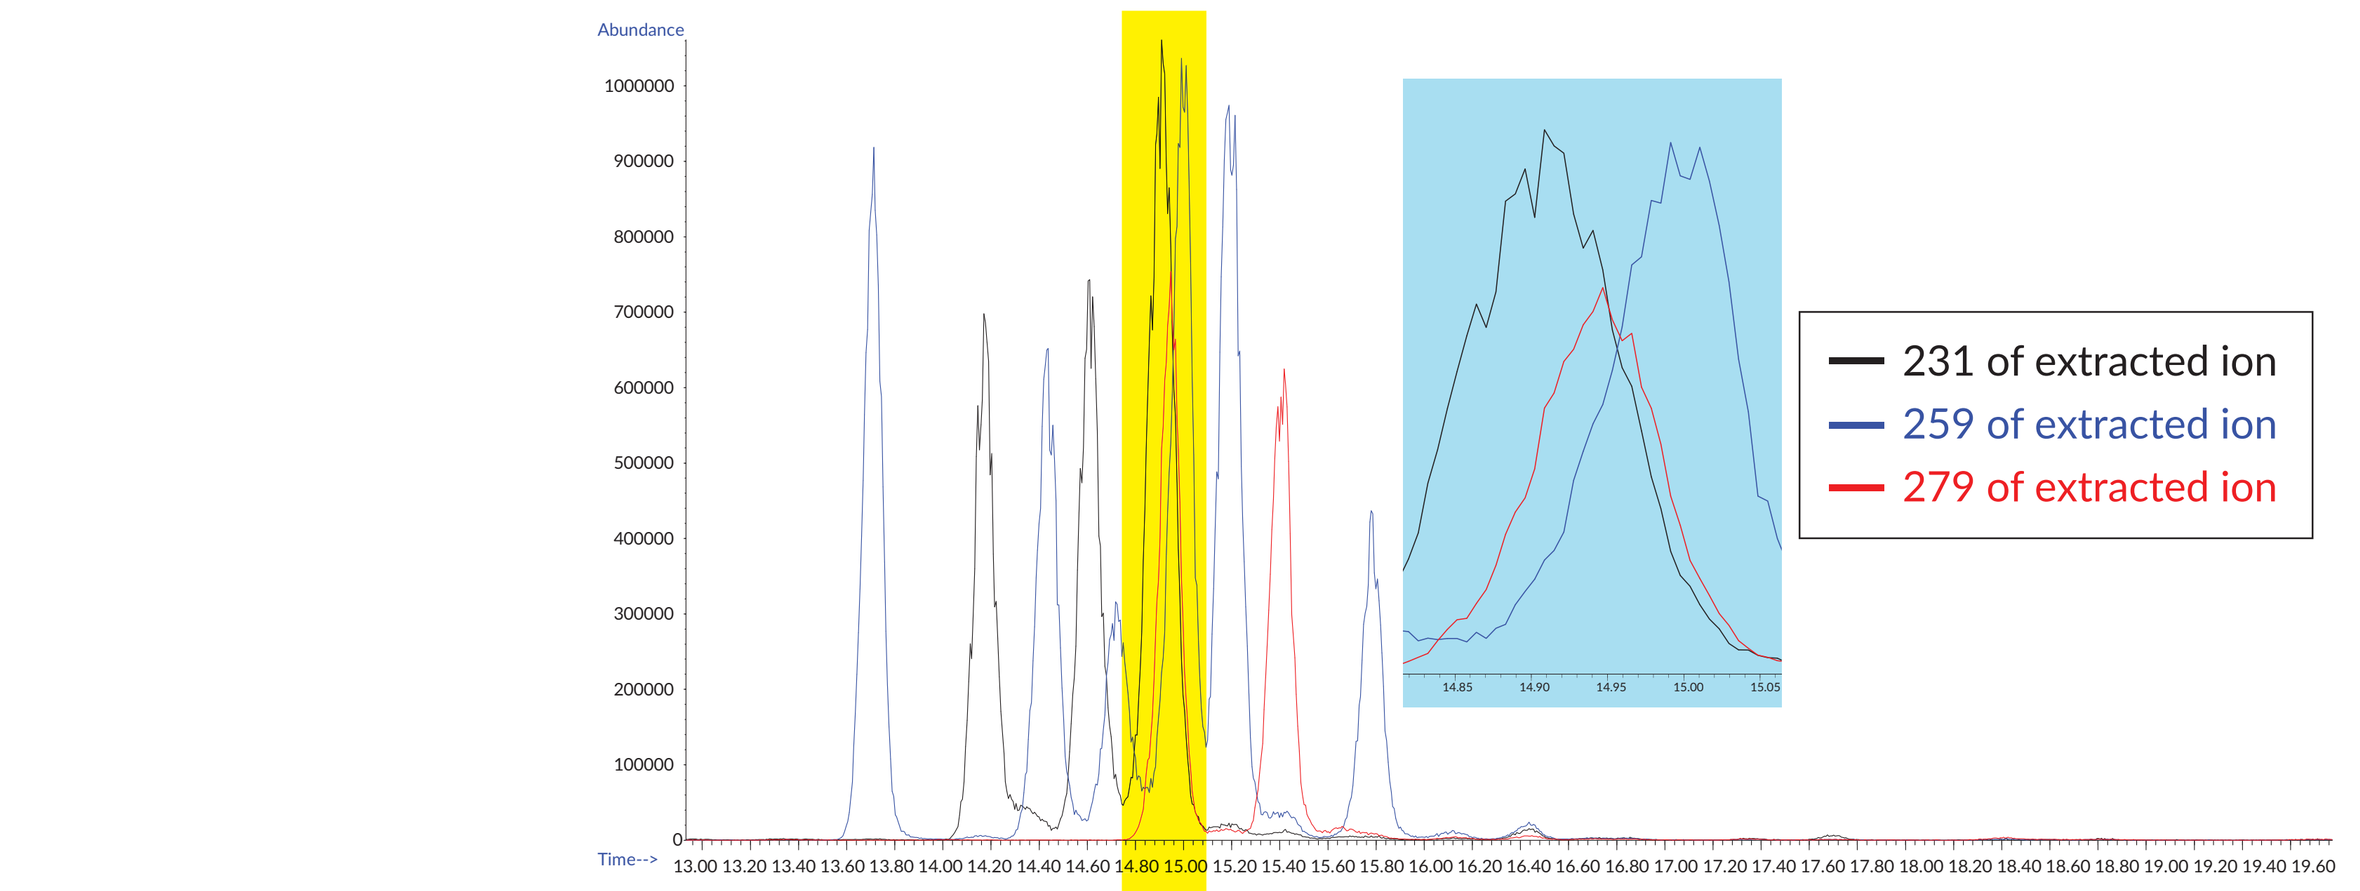
231 of extracted ion
259 of extracted ion
279 of extracted ion

Figure 3. EIC of *m/z* 231, 259, and 279.

As reported in Tables 2 and 3, the fragment ions 231 and 279 are the base peaks of methyl acetyl fentanyl and fluoro-methoxyacetyl fentanyl groups, respectively. Fragment ion 259 is the base peak of the methyl thiofentanyl and methylfentanyl analog groups. The second and third most abundant ion peaks for each of the three analog groups were reported in **Table 2** and **Table 3**. Secondary EICs (**Figure 4**) comprised of the key ion fragments and the retention time further narrowed down the identity of unknown compounds A, B, and C.

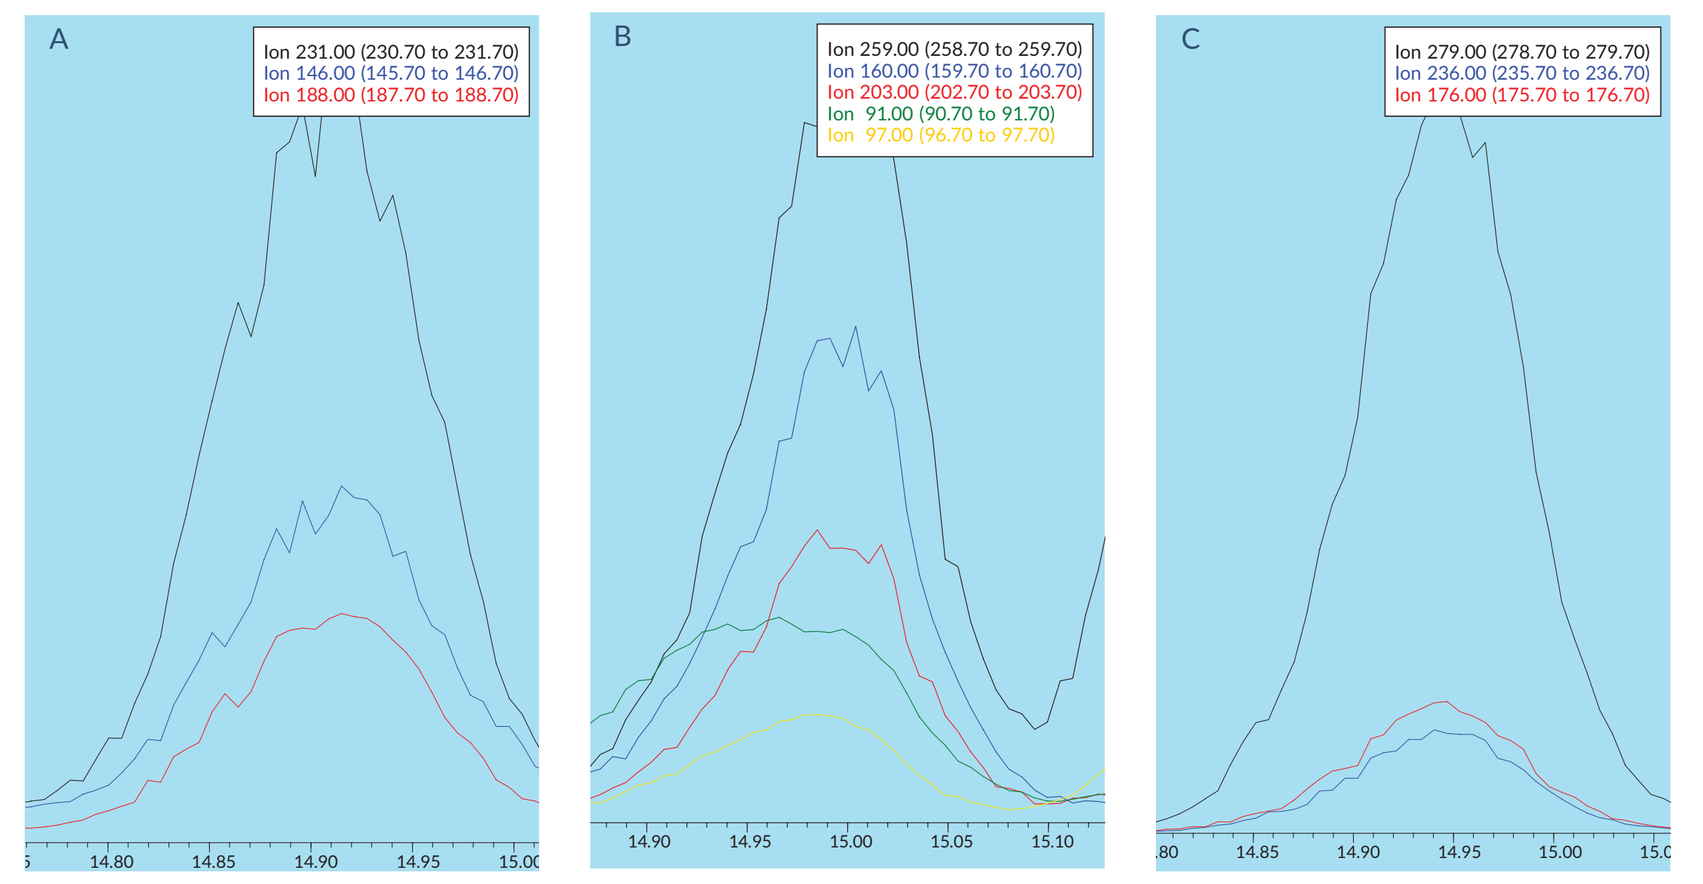
A
Ion 231.00 (230.70 to 231.70)
Ion 146.00 (145.70 to 146.70)
Ion 188.00 (187.70 to 188.70)
B
Ion 259.00 (258.70 to 259.70)
Ion 160.00 (159.70 to 160.70)
Ion 203.00 (202.70 to 203.70)
Ion 91.00 (90.70 to 91.70)
Ion 97.00 (96.70 to 97.70)
C
Ion 279.00 (278.70 to 279.70)
Ion 236.00 (235.70 to 236.70)
Ion 176.00 (175.70 to 176.70)

Figure 4. Expanded EIC of **A.** *m/z* 231, 146, 188, T = 14.915 min, **B.** *m/z* 259, 160, 203, 91, 97, T = 15.013 min, and **C.** *m/z* 279, 236, 176, T = 14.945 min

The RRT for unknown B was calculated. The retention time of the reference was determined to be 13.389; the calculated RRT for unknown B is 1.121. Referring to **Table 3**, (±)-*cis*-3-methyl thiofentanyl has an RRT of 1.122 and *ortho*-methylfentanyl has an RRT = 1.125. Fragment ions at *m/z* 91 and 97 further support the coelution of the two components. The 1 mg/ml concentration is suspected to be the cause of the coelution of the two components; resolution may be obtained by diluting the sample and reanalyzing.

Tuning Comparison

To explore whether the fragmentation of the isobaric compounds cyclopropyl fentanyl and crotonyl fentanyl could be differentiated by altering the tuning parameters, the mass spectra (MS) of both items were analyzed using Standard Spectra Autotune (S-tune) and Autotune (A-tune). The MS of both compounds slightly changed when analyzed under the different tuning parameters (**Table 5**). However, the two isomers could not be differentiated when using the same tuning type (**Figure 5**).

Table 5. Tune parameters for tuning type comparison

	A-tune	S-tune
69 Abundance Target, Counts	430,976	813,312
Mass 219 %:	101.84	59.64
Mass 502 %:	2.21	2.34

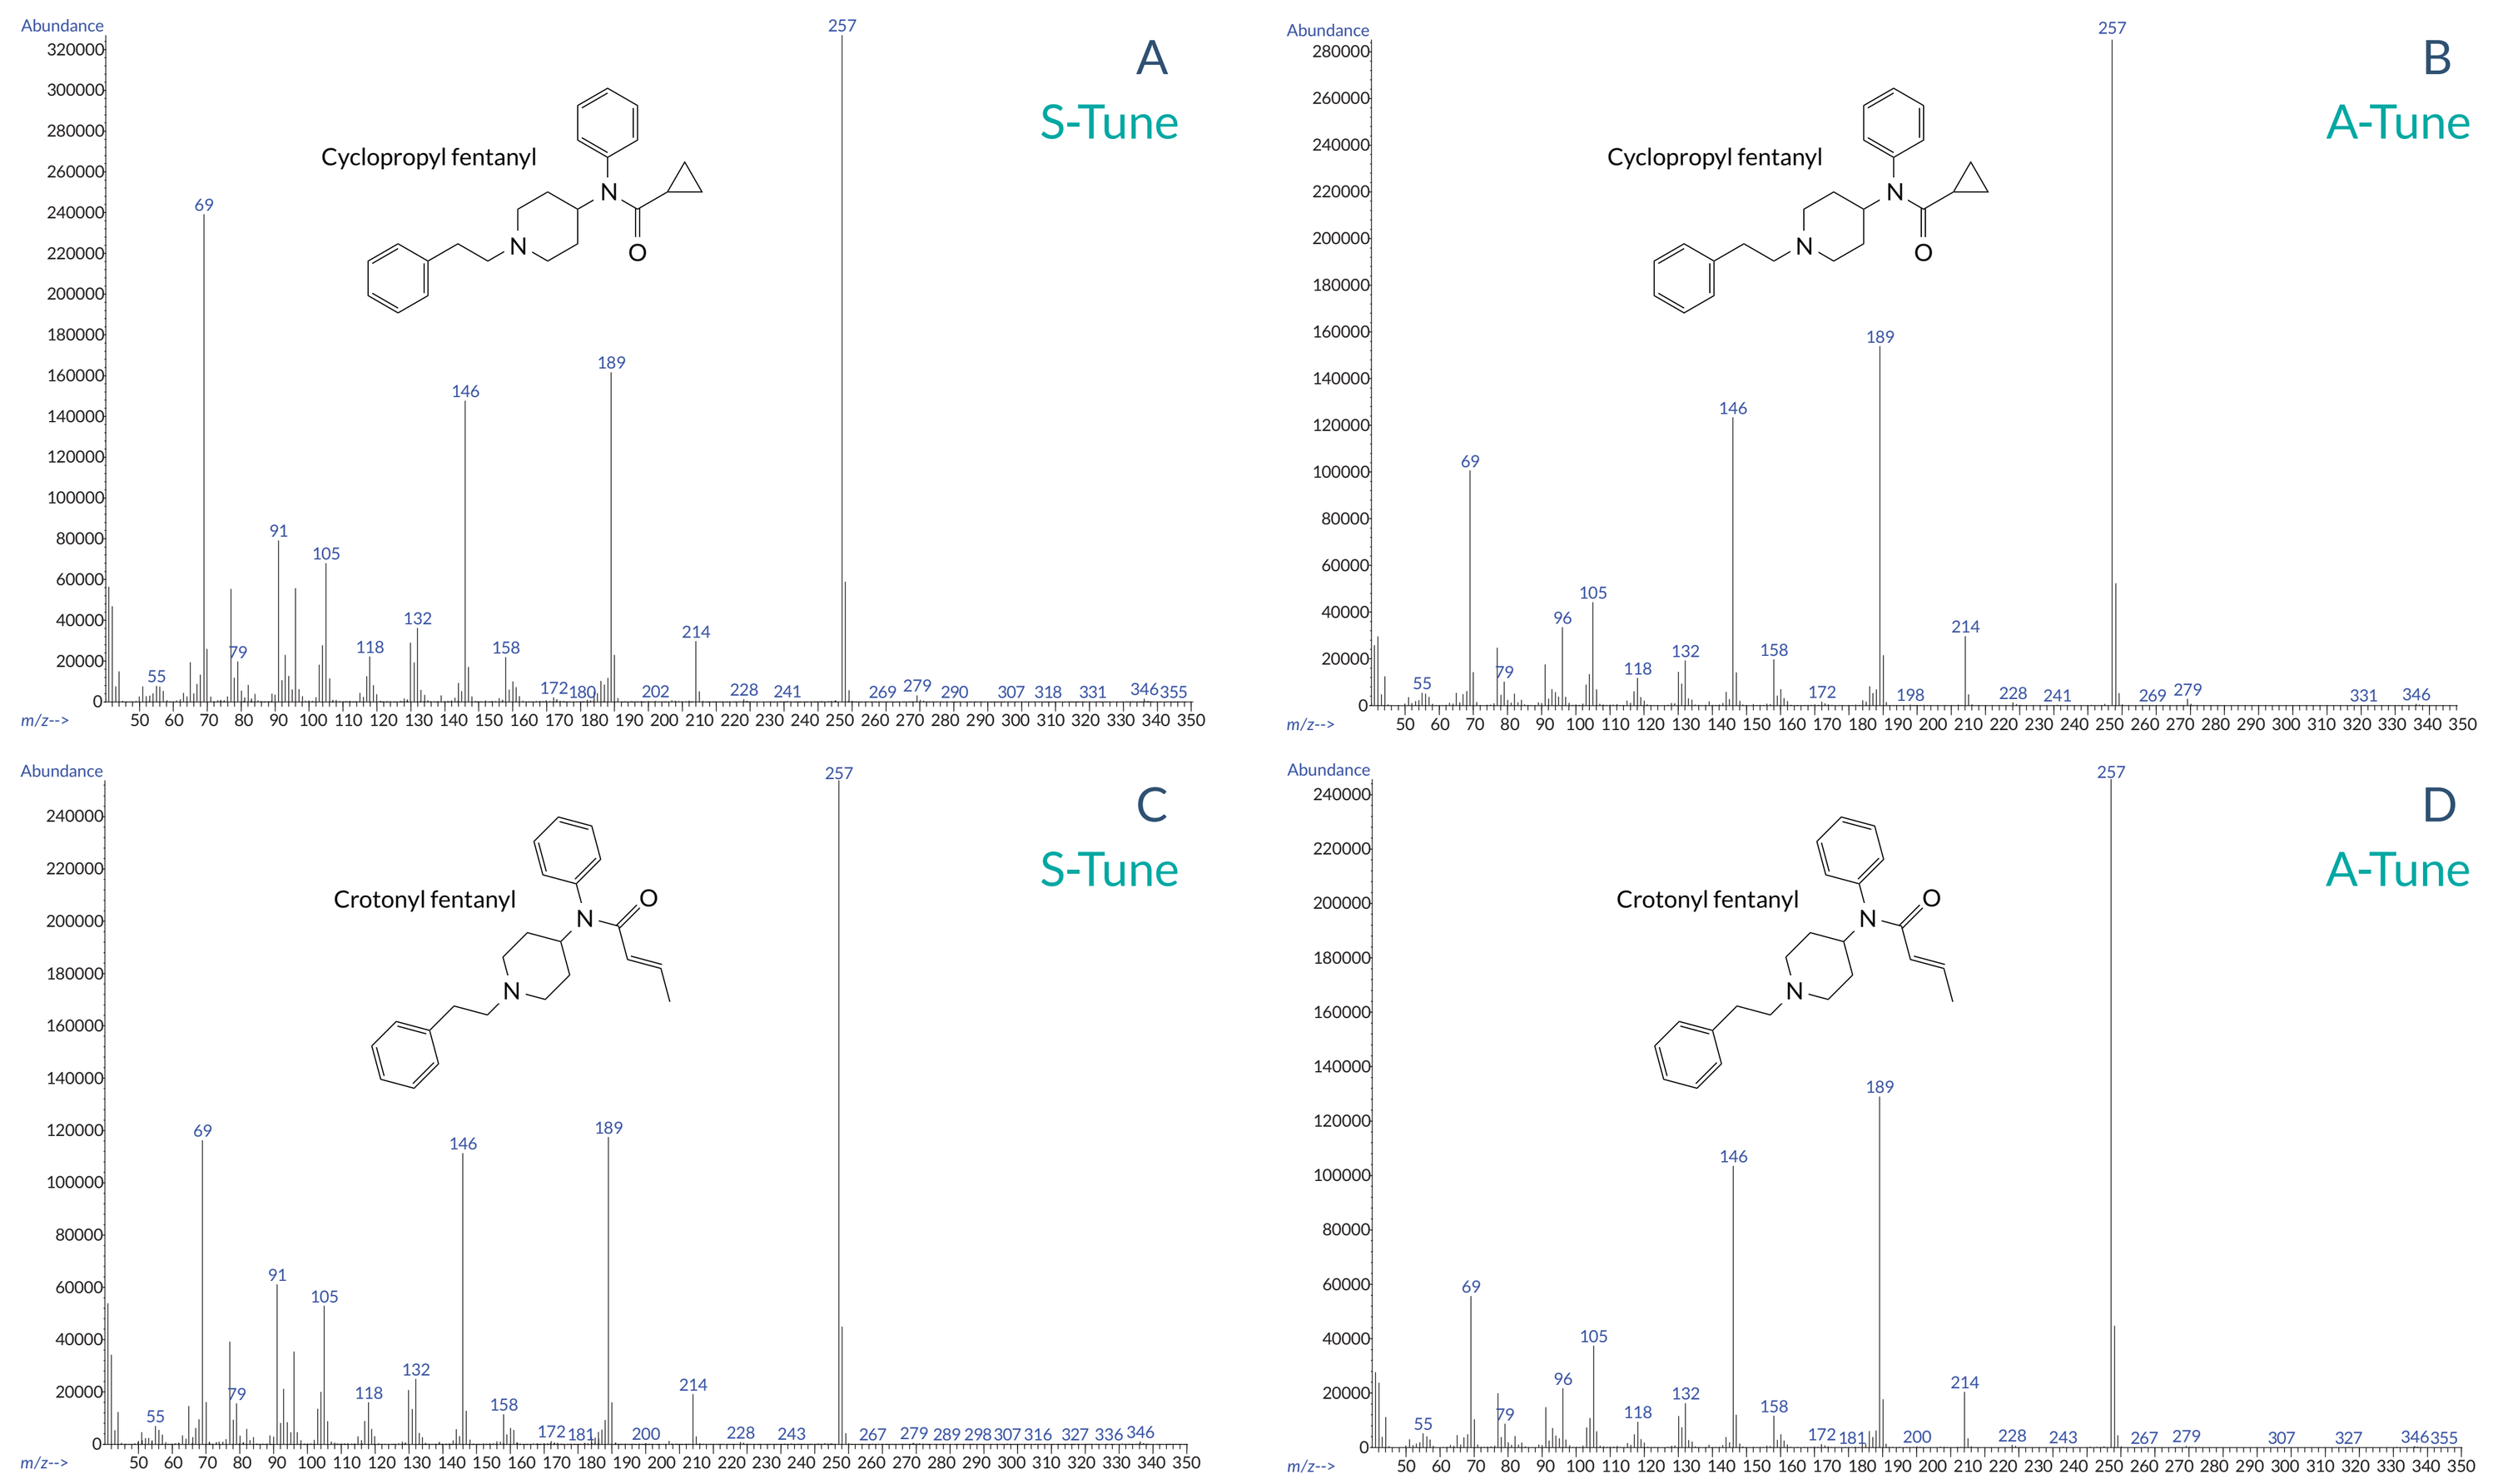
A
S-Tune
Cyclopropyl fentanyl
B
A-Tune
Cyclopropyl fentanyl
C
S-Tune
Crotonyl fentanyl
D
A-Tune
Crotonyl fentanyl

Figure 5. Cyclopropyl fentanyl with **A.** S-tune and **B.** A-tune; Crotonyl fentanyl with **C.** S-tune and **D.** A-tune.

Conclusion

- Certain isomers may be differentiated using the combination of GC separation and fragmentation.
- EIC may be a useful tool to differentiate the fentanyl analogs in a complex mixture where analyte separation has not been fully attained.
- When using EIC, selecting several ion fragments provides higher confidence for confirming identity of an unknown.
- Tuning type slightly affected the ratio of the fragments but did not contribute to a significant difference in the examined isobaric materials.

Future Work

1. Continue efforts with developing GC-MS methods for isomer differentiation of new and relevant forensic substances.
2. Continue similar isomer separation studies using liquid chromatography with diode-array detector (LC-DAD) for the 60 fentanyl analogs in this study.

Any future work, literature, or posters will be shared through Cayman Chemical's website, www.caymanchem.com.

Acknowledgement

Special thanks to Rob Schelkun for his structure drawing expertise and all our colleagues in the Forensic Chemistry Division for their review and feedback on the content.

References

1. Pierzynski, H.G., Liu, J., Miller, M., *et al.*, Methods to differentiate base peak 257 fentanyls: Methacrylfentanyl, cyclopropyl fentanyl, and crotonyl fentanyl (Application Note) (2018). Retrieved from Cayman Chemical website: https://www.caymanchem.com/Literature/Methods to Differentiate Base Peak 257 Fentanyls
2. Mallette, J.R., Casale, J.F., and Hays, P.A. Characterization and differentiation of cyclopropylfentanyl from E-crotonylfentanyl, Z-crotonylfentanyl, and 3-butenylfentanyl. *Sci. Justice* **59(1)**, 67-74 (2019).
3. Mallette, J.R., Casale, J.F., Toske, S.G., *et al.* Characterization of (2R,4S)- and (2R,4R)-2-methylfentanyl and their differentiation from *cis*- and *trans*-3-methylfentanyl. *Forensic Chem.* **8**, 64-71 (2018).
4. Pierzynski, H.G., Newbauer, L., Choi, C., *et al.*, Tips for interpreting GC-MS fragmentation of unknown substituted fentanyls, *Cayman Currents*, **28(293)** Fall 2017. Retrieved from Cayman Chemical website: https://www.caymanchem.com/Literature/%E2%80%8Bcayman-currents-issue-28-fentanyl-identification